Nonclinical Data for PGN-EDODM1 Demonstrated Mechanistic and Meaningful Activity for the Potential Treatment of DM1

Shaoxia Yu[1], Arnaud Klein[2],

Jeffrey Foy[1], Denis Furling[2], Jane Larkindale

[1]PepGen Inc., Boston, MA, USA, [2]Sorbonne Université, Inserm, Institut de Myologie, Centre de Recherche en Myologie, Paris, France

INTRODUCTION

The **Enhanced Delivery Oligonucleotide (EDO)** platform is **engineered to optimize the tissue penetration, cellular uptake and nuclear delivery** of oligonucleotide therapeutic candidates.

Myotonic dystrophy type 1 (DM1) is a multi-systemic disease that has a **significant impact on quality of life.** There are **no approved therapies** that address the underlying cause of DM1.

Limited delivery and distribution of unconjugated oligonucleotides to affected tissues limit their potential effectiveness in DM1.

PGN-EDODM1 is a proprietary peptide-conjugated PMO (PPMO) and was evaluated in multiple nonclinical models, including DM1 human-derived muscle cells, the HSA^{LR} mouse model of DM1, wild-type (WT) mice, and non-human primates (NHPs).

PGN-EDODM1 is an investigational EDO and is being evaluated in people with DM1 in a Phase 1 single ascending dose study (FREEDOM) and a Phase 2 multiple ascending dose study (FREEDOM2).

MECHANISM OF PGN-EDODM1

- **PGN-EDODM1** is engineered to bind selectively to the pathogenic CUG repeat expansion present in *DMPK* transcripts.
- This reduces the ability of these CUG repeats to form hairpin loops and sequester RNA splicing proteins, including MBNL1.
- Liberated MBNL1 restores correct splicing.

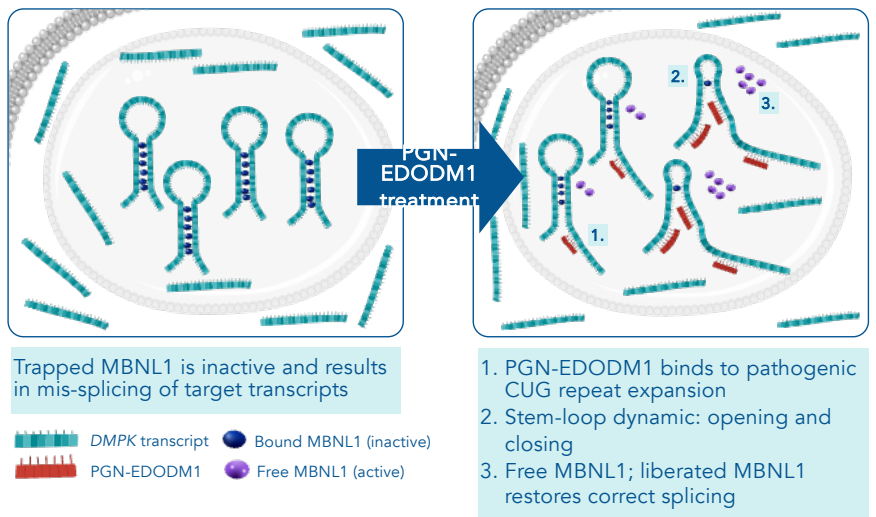

PGN-EDODM1 treatment
2.
3.
1.
Trapped MBNL1 is inactive and results in mis-splicing of target transcripts
DMPK transcript
Bound MBNL1 (inactive)
PGN-EDODM1
Free MBNL1 (active)

1. PGN-EDODM1 binds to pathogenic CUG repeat expansion
2. Stem-loop dynamic: opening and closing
3. Free MBNL1; liberated MBNL1 restores correct splicing

CELLULAR DELIVERY AND ACTIVITY DATA

EDO TECHNOLOGY INCREASED CELLULAR UPTAKE AND NUCLEAR DELIVERY OF OLIGONUCLEOTIDE THERAPEUTICS IN MUSCLE CELLS

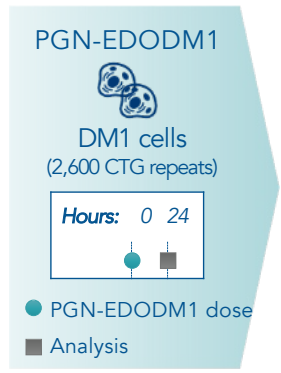

PGN-EDODM1
DM1 cells
(2,600 CTG repeats)
Hours: 0 24
PGN-EDODM1 dose
Analysis

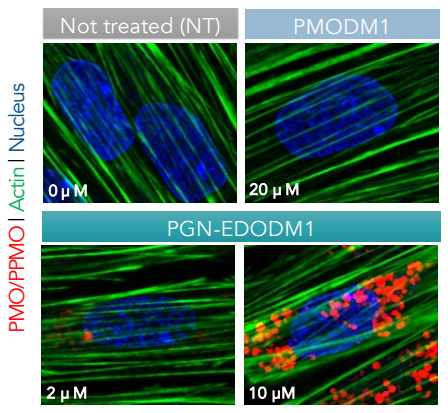

Not treated (NT)
PMODM1
PGN-EDODM1
PMO/PPMO | Actin | Nucleus
0 μM
20 μM
2 μM
10 μM

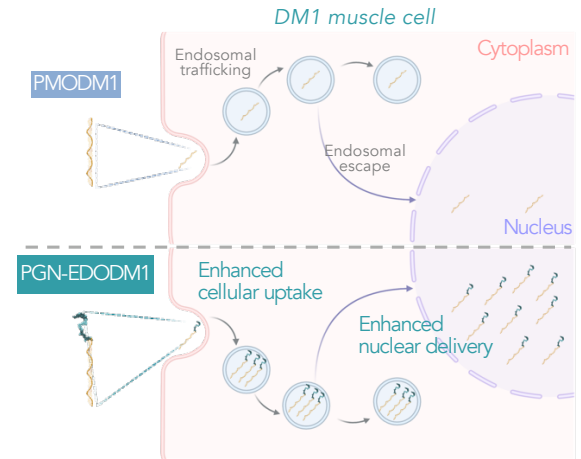

DM1 muscle cell
Cytoplasm
PMODM1
Endosomal trafficking
Endosomal escape
Nucleus
PGN-EDODM1
Enhanced cellular uptake
Enhanced nuclear delivery

EDO ENHANCED CELLULAR AND NUCLEAR UPTAKE

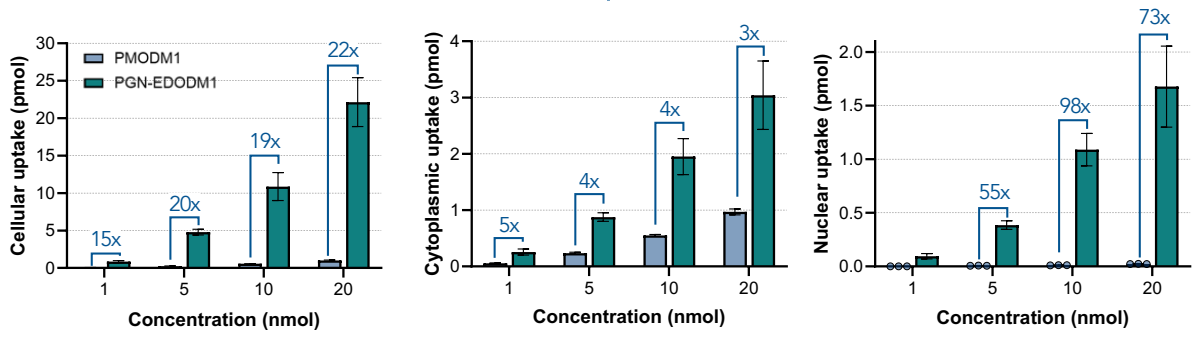

Total cell
Cytoplasm
Nucleus
PMODM1
PGN-EDODM1
Cellular uptake (pmol)
Cytoplasmic uptake (pmol)
Nuclear uptake (pmol)
15x
20x
19x
22x
5x
4x
4x
3x
55x
98x
73x
Concentration (nmol)

PGN-EDODM1 REDUCED TOXIC FOCI, LIBERATED MBNL1, CORRECTED MIS-SPLICING AND INCREASED MBNL1 PROTEIN LEVELS IN DM1 MUSCLE CELLS

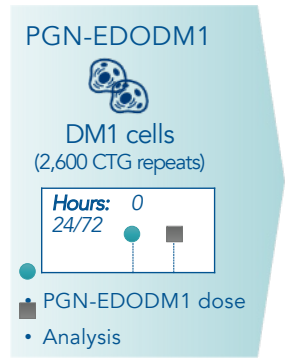

PGN-EDODM1
DM1 cells
(2,600 CTG repeats)
Hours: 0 24/72
PGN-EDODM1 dose
Analysis

TOXIC FOCI REDUCTION

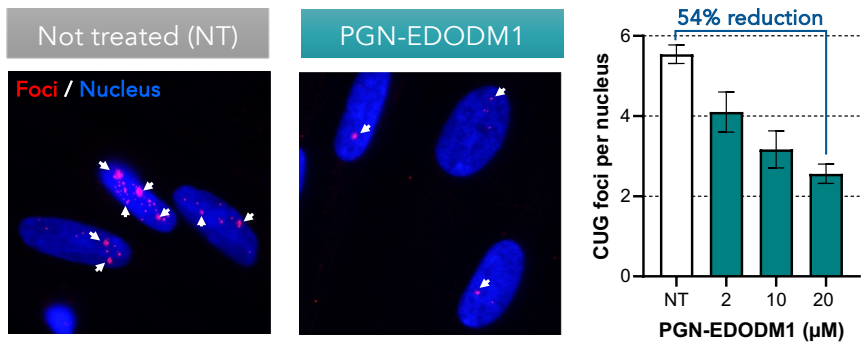

Not treated (NT)
PGN-EDODM1
Foci / Nucleus
54% reduction
CUG foci per nucleus
NT
2
10
20
PGN-EDODM1 (μM)

MBNL1 LIBERATION

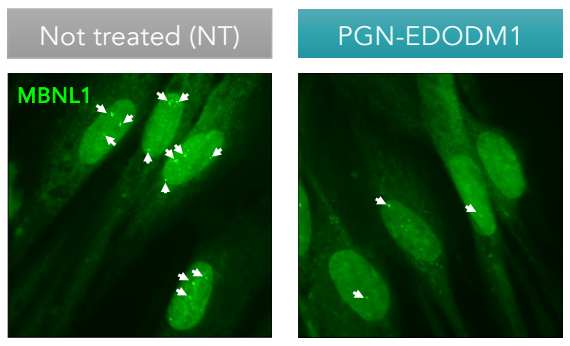

Not treated (NT)
PGN-EDODM1
MBNL1

MIS-SPLICING CORRECTION

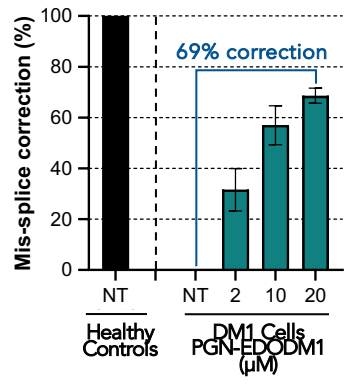

Across multiple transcripts
69% correction
Mis-splice correction (%)
NT
NT
2
10
20
Healthy Controls
DM1 Cells PGN-EDODM1 (μM)

MBNL1 PROTEIN INCREASE

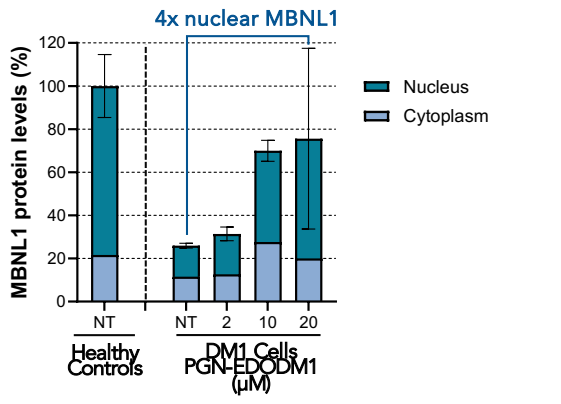

4x nuclear MBNL1
Nucleus
Cytoplasm
MBNL1 protein levels (%)
NT
NT
2
10
20
Healthy Controls
DM1 Cells PGN-EDODM1 (μM)

HSA^{LR} MOUSE MODEL AND NON-HUMAN PRIMATE (NHP) DATA

REPEAT DOSING OF PGN-EDODM1 IN HSA^{LR} MICE ENHANCED CORRECTION OF MIS-SPLICING, REVERSED MYOTONIA AND INCREASED MUSCLE DELIVERY

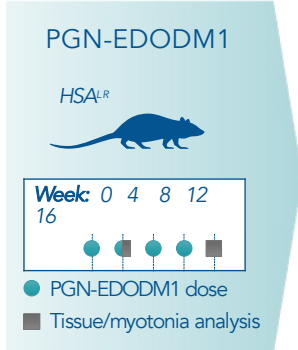

PGN-EDODM1
HSA LR
Week: 0 4 8 12 16
PGN-EDODM1 dose
Tissue/myotonia analysis

TISSUE CONCENTRATION

Skeletal muscle

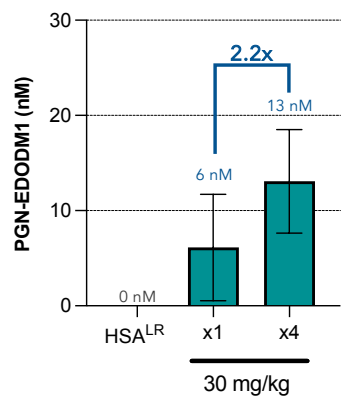

2.2x
13 nM
6 nM
0 nM
PGN-EDODM1 (nM)
HSA LR
x1
x4
30 mg/kg

MIS-SPLICING CORRECTION

Across multiple transcripts

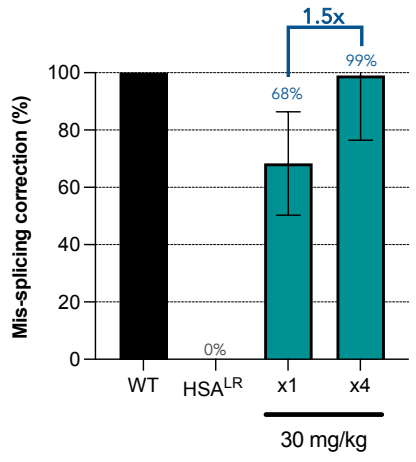

1.5x
99%
68%
0%
Mis-splicing correction (%)
WT
HSA LR
x1
x4
30 mg/kg

REVERSAL OF MYOTONIA

Action myotonia

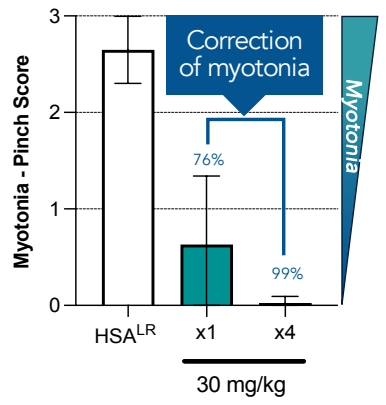

Correction of myotonia
Myotonia
76%
99%
Myotonia - Pinch Score
HSA LR
x1
x4
30 mg/kg

Increased levels of PGN-EDODM1 in tissue with repeat dosing | **99% correction** across multiple transcripts | **Complete correction of myotonia** observed after repeat dosing

FAVORABLE SAFETY PROFILE IN NHPs SUPPORTED PROGRESSION TO CLINICAL STUDIES

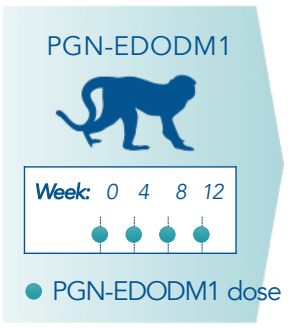

PGN-EDODM1
Week: 0 4 8 12
PGN-EDODM1 dose

SERUM CREATININE

60 mg/kg

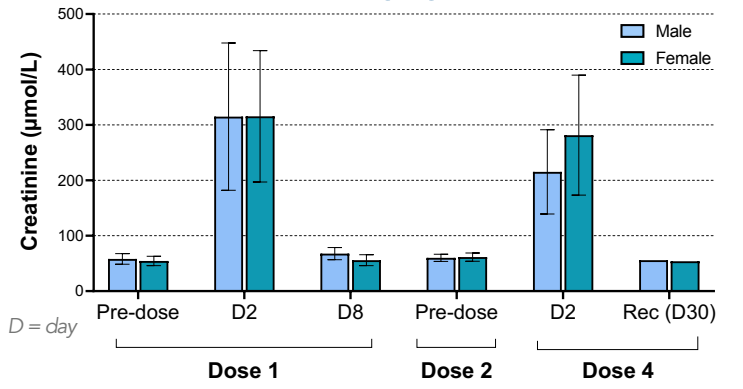

Male
Female
Creatinine (μmol/L)
D = day
Pre-dose
D2
D8
Pre-dose
D2
Rec (D30)
Dose 1
Dose 2
Dose 4

- Non-adverse transient increases in serum creatinine were observed at 60 mg/kg and resolved within a week postdose and did not worsen with repeat dosing.
- No adverse findings in the kidney at 60 mg/kg (Q4W; 4 doses).
- No notable hematologic or hepatic effects, no cardiovascular effects.

CONCLUSIONS & NEXT STEPS

- **PGN-EDODM1** was engineered **to bind to the pathogenic CUG repeat expansion** present in *DMPK* transcripts in people with DM1 and is not intended to degrade *DMPK*.
- Treatment with **PGN-EDODM1** resulted in **nuclear delivery**, **reduction of toxic foci, liberation of MBNL1, correction of mis-splicing,** and **increased MBNL1 protein levels** in DM1 human-derived muscle cells.
- In the HSA^{LR} DM1 mouse model, **robust mis-splicing correction and reversal of myotonia** were observed with a single 30 mg/kg dose; durable mis-splicing correction was observed **through 24 weeks.**
- **Increased levels of tissue delivery, enhanced mis-splicing correction** and **reversal of myotonia** were observed with repeat dosing in HSA^{LR} mice.
- **PGN-EDODM1 was well tolerated in NHP GLP repeat-dose toxicity studies at 60 mg/kg; repeat dosing did not exacerbate increases in serum creatinine.**
- **FREEDOM-DM1 Phase 1** randomized, double-blind, placebo-controlled **single ascending dose study in people with DM1** is **enrolling in Canada,** the **UK** and the **US. FREEDOM2 Phase 2** randomized, double-blind, placebo-controlled **multiple ascending dose study in people with DM1** is **enrolling in Canada** and the **UK.**
- **Nonclinical data** in DM1 cells, HSA^{LR} mice and NHP support the **development of PGN-EDODM1** and the **FREEDOM1-DM1 Phase 1 and FREEDOM2-DM1 Phase 2 clinical studies**.

Disclosures: [1]Employees of PepGen Inc. and hold PepGen equity; [2]Work conducted under a sponsored research agreement